CHARTING A COMPLEX WORLD.

TECHATLAS | PARKINSON'S DISEASE

A DIVISION OF RACAPITAL

MAY 2017

OUT OF DATE. DO NOT RELY UPON.

TECHATLAS STANDARD OF CARE

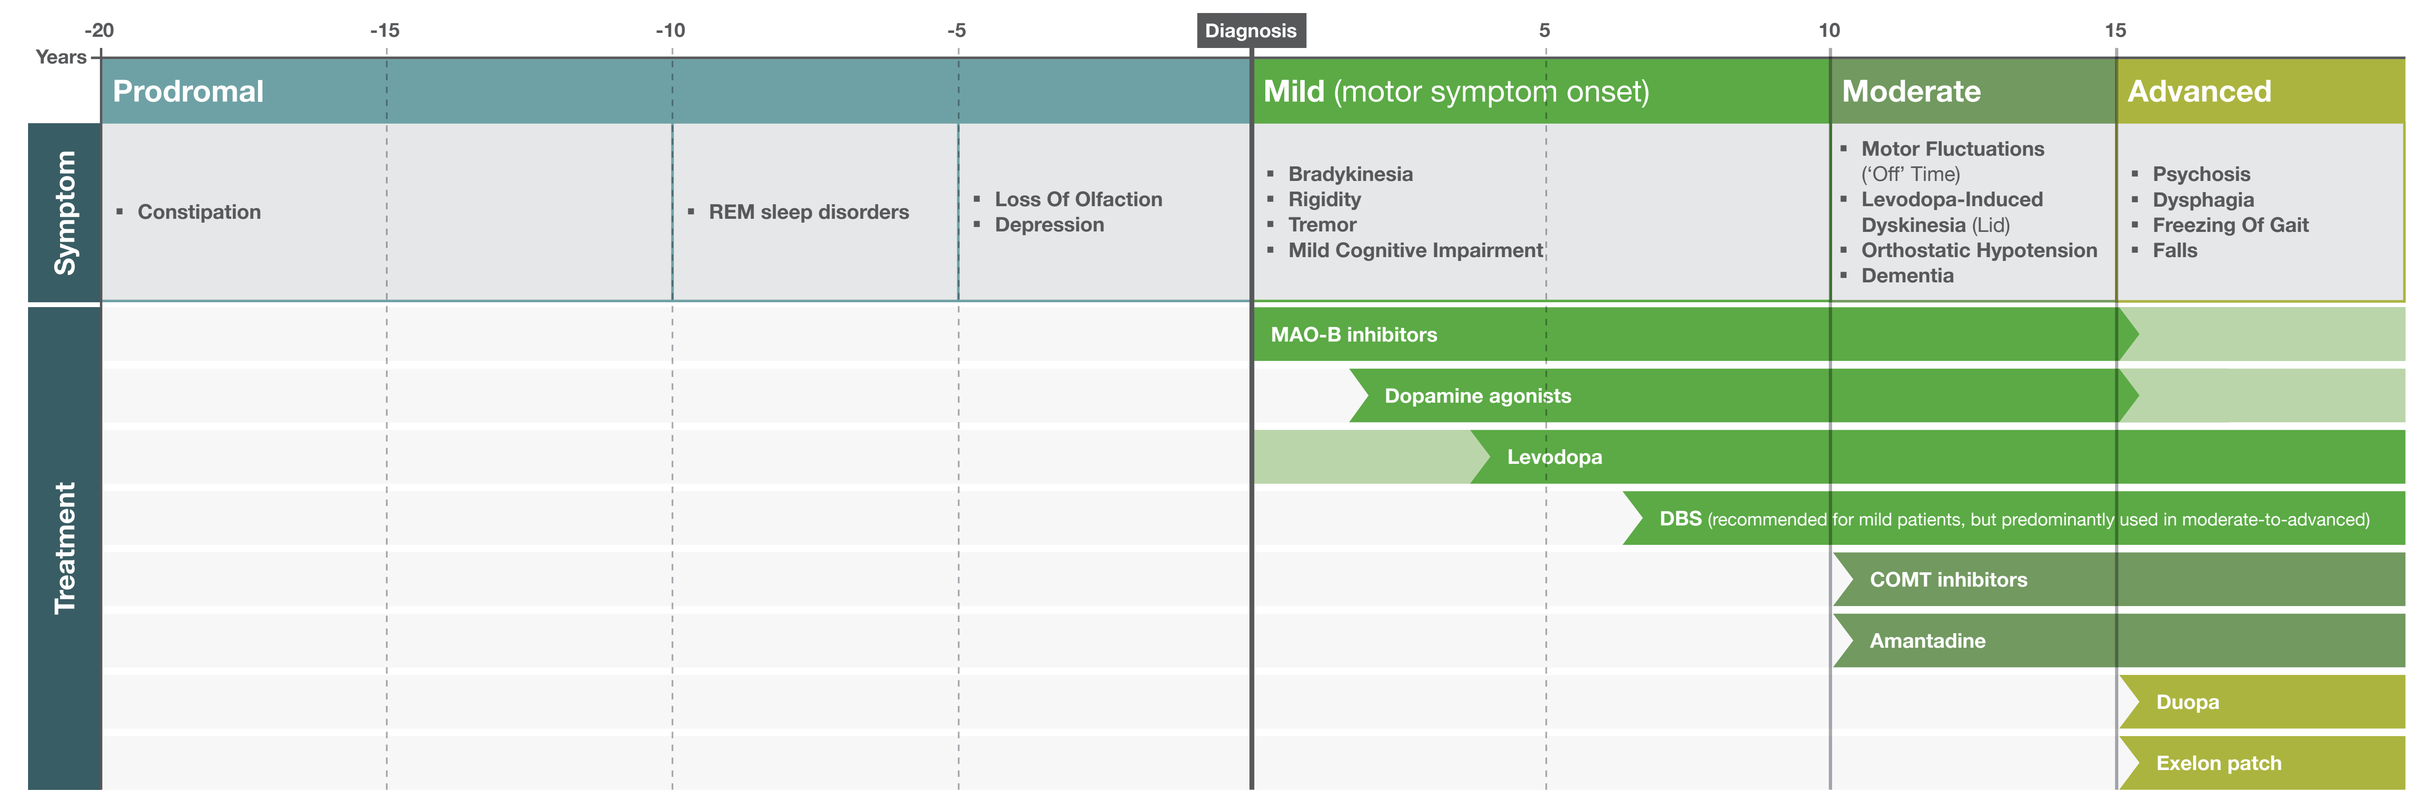

Years
-20
-15
-10
-5
Diagnosis
5
10
15
Prodromal
Mild (motor symptom onset)
Moderate
Advanced
Symptom
Constipation
REM sleep disorders
Loss Of Olfaction
Depression
Bradykinesia
Rigidity
Tremor
Mild Cognitive Impairment
Motor Fluctuations ('Off' Time)
Levodopa-Induced Dyskinesia (Lid)
Orthostatic Hypotension
Dementia
Psychosis
Dysphagia
Freezing Of Gait
Falls
Treatment
MAO-B inhibitors
Dopamine agonists
Levodopa
DBS (recommended for mild patients, but predominantly used in moderate-to-advanced)
COMT inhibitors
Amantadine
Duopa
Exelon patch

2014 WW SALES OF MARKETED PD THERAPIES

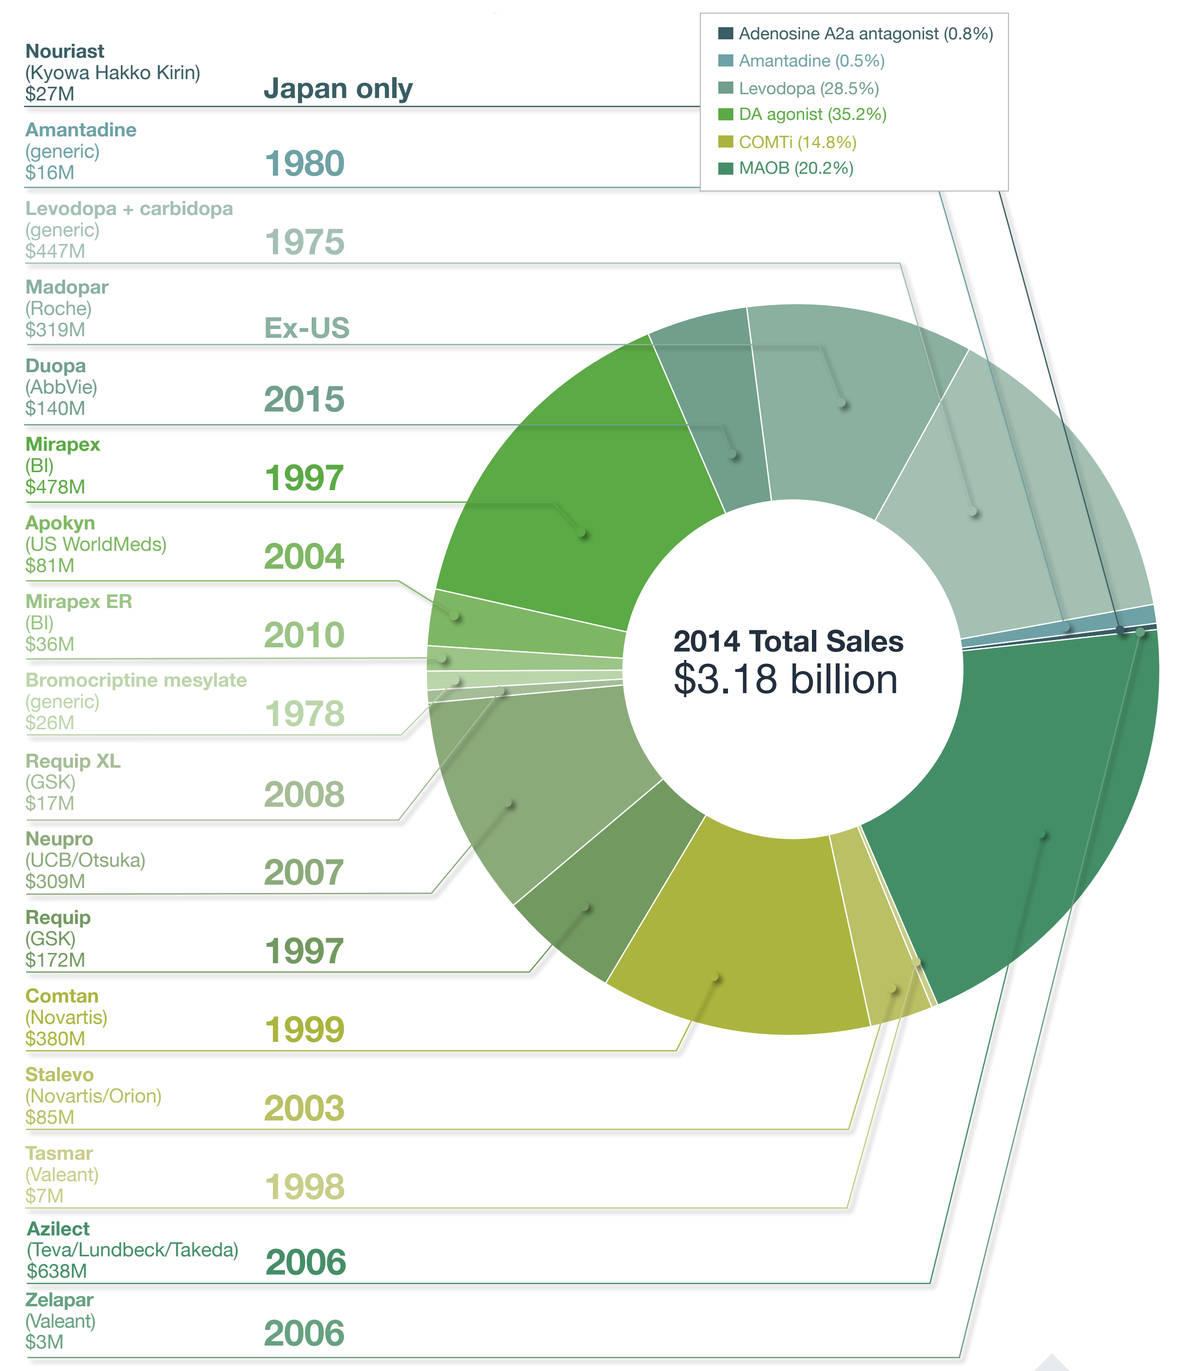

Adenosine A2a antagonist (0.8%)
Amantadine (0.5%)
Levodopa (28.5%)
DA agonist (35.2%)
COMTI (14.8%)
MAOB (20.2%)
2014 Total Sales
$3.18 billion
Nouriast (Kyowa Hakko Kirin) $27M — Japan only
Amantadine (generic) $16M — 1980
Levodopa + carbidopa (generic) $447M — 1975
Madopar (Roche) $319M — Ex-US
Duopa (AbbVie) $140M — 2015
Mirapex (BI) $478M — 1997
Apokyn (US WorldMeds) $81M — 2004
Mirapex ER (BI) $36M — 2010
Bromocriptine mesylate (generic) $26M — 1978
Requip XL (GSK) $17M — 2008
Neupro (UCB/Otsuka) $309M — 2007
Requip (GSK) $172M — 1997
Comtan (Novartis) $380M — 1999
Stalevo (Novartis/Orion) $85M — 2003
Tasmar (Valeant) $7M — 1998
Azilect (Teva/Lundbeck/Takeda) $638M — 2006
Zelapar (Valeant) $3M — 2006

LEVODOPA PHARMACOKINETICS

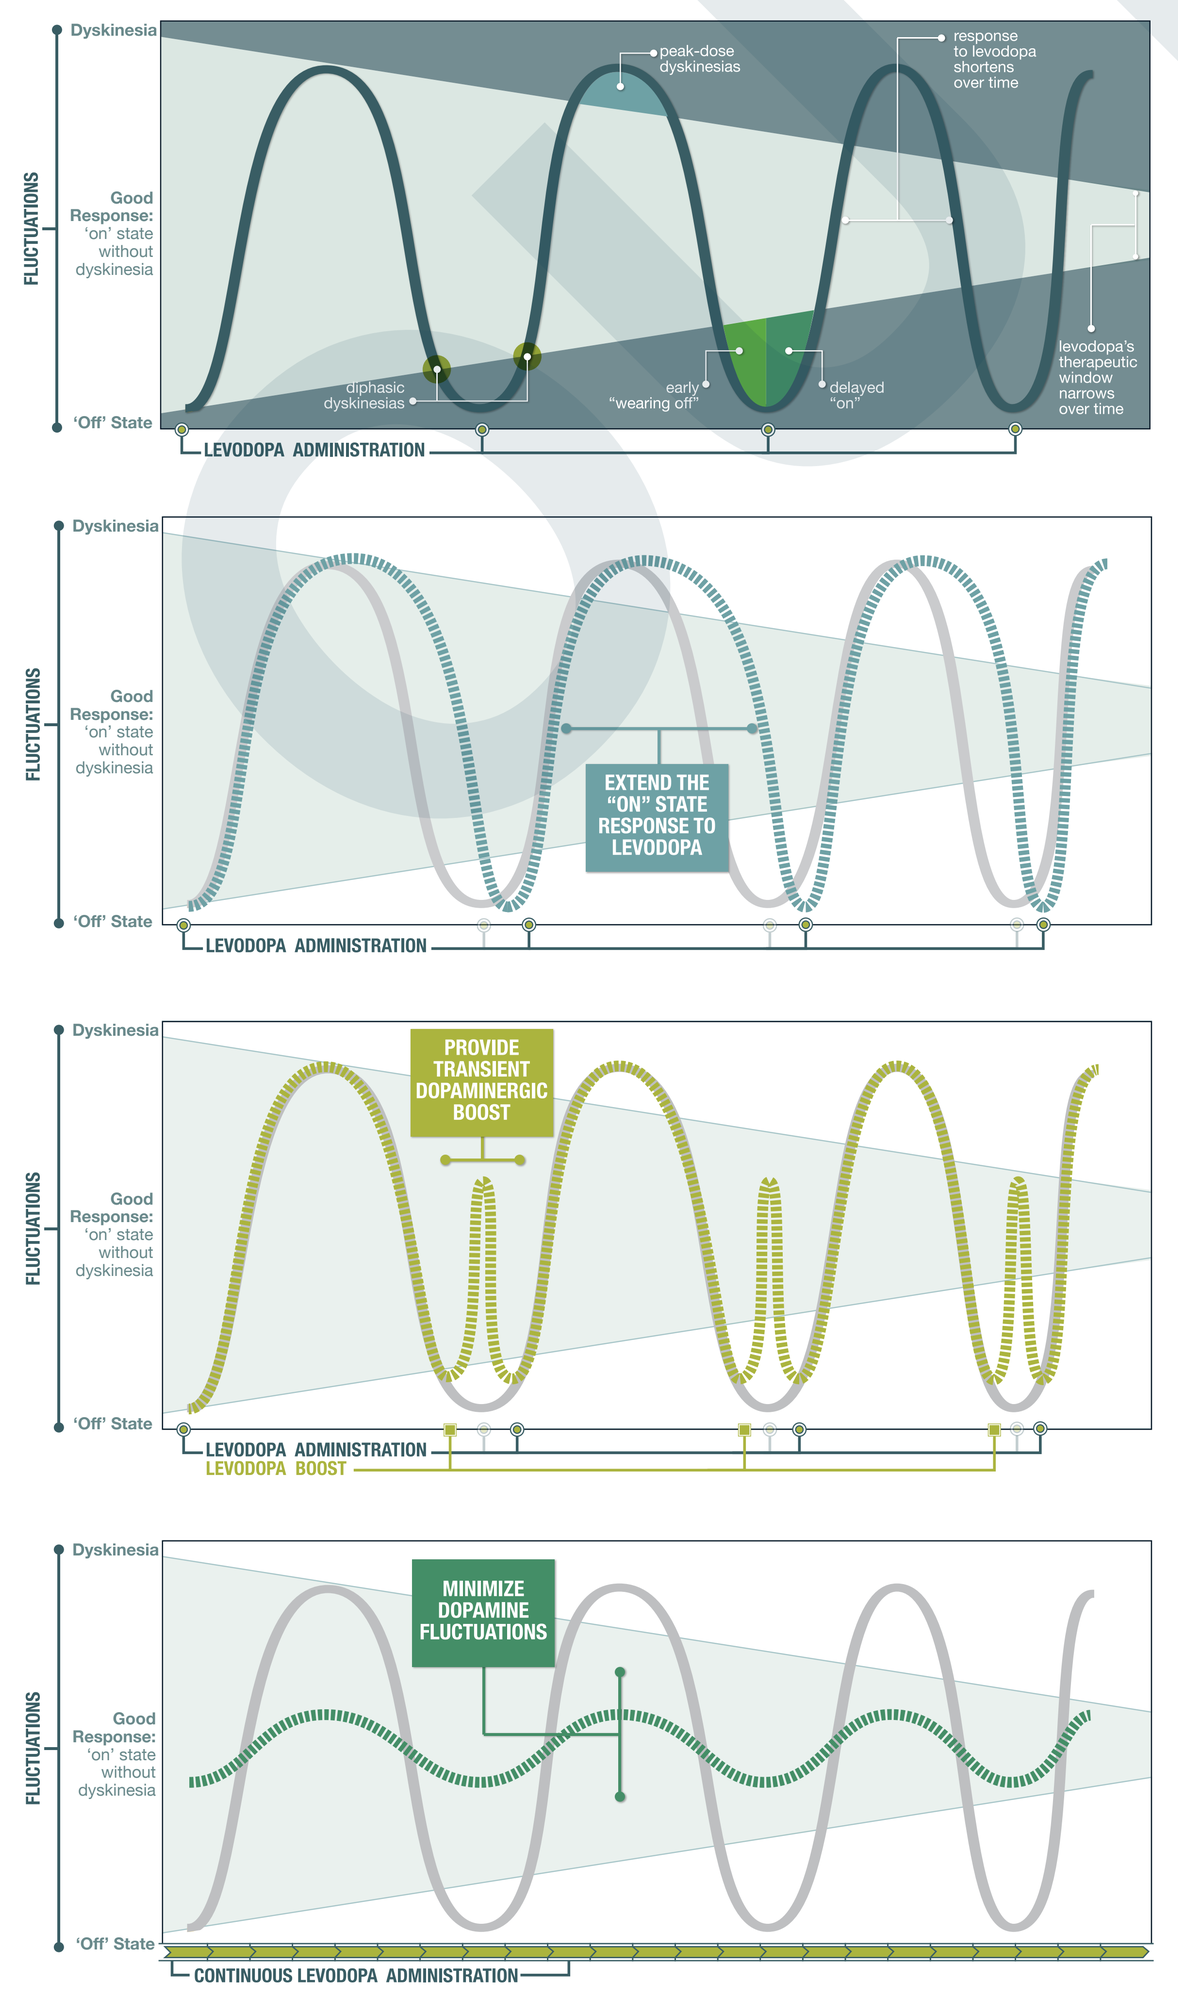

FLUCTUATIONS
Dyskinesia
Good Response: 'on' state without dyskinesia
'Off' State
peak-dose dyskinesias
response to levodopa shortens over time
diphasic dyskinesias
early "wearing off"
delayed "on"
levodopa's therapeutic window narrows over time
LEVODOPA ADMINISTRATION
EXTEND THE "ON" STATE RESPONSE TO LEVODOPA
PROVIDE TRANSIENT DOPAMINERGIC BOOST
LEVODOPA BOOST
MINIMIZE DOPAMINE FLUCTUATIONS
CONTINUOUS LEVODOPA ADMINISTRATION

Objective: Manage Symptoms

In the absence of disease-modifying therapies, Parkinson's disease (PD) treatment remains centered on replacing lost dopaminergic signaling. Strategies include supplying the dopamine pro-drug L-DOPA (levodopa), using small molecule dopamine receptor agonists, and inhibiting dopamine metabolism with MAO-B inhibitors. Non-dopaminergic therapies are also used, but with more limited success.

Manage primary motor symptoms

Cardinal motor symptoms are resting tremor, bradykinesia (slowness of movement), rigidity, and gait disturbance/postural instability. Levodopa remains the best therapy for motor symptoms, but its use is associated with long-term side effects including levodopa-induced dyskinesia (LID) and end-of-dose wearing off ('off' time).

Noninvasive dopaminergic therapies

Basal levodopa — Most common long-term treatment; levodopa is the main therapy for nearly all patients as the disease progresses.

- **Gold standard: immediate-release levodopa formulations** — Levodopa is co-formulated with a peripheral decarboxylase inhibitor (carbidopa in the US) to inhibit plasma levodopa metabolism, which allows adequate concentrations of levodopa to reach the brain.
 - G (Sinemet/carbidopa+levodopa)
 - M Roche (Madopar/benserazide+levodopa)
 - M Vernalis (Sirio/carbidopa+melevodopa)
- **Extended-release levodopa formulations**
 - **Formulations with immediate release component** — Faster time to 'on'
 - M Impax (Rytary/carbidopa+levodopa)
 - RCT: 4Q17 ••• Intec Pharma (AP-CDLD/carbidopa+levodopa accordion pill)
 - RCT: 4Q17 •• Impax (IPX203)
 - **Formulations without immediate release component** — Slower time to 'on', only used for bedtime dose
 - G (Sinemet CR/carbidopa+levodopa CR)
 - M Roche (Madopar CR/benserazide+levodopa)

Other dopaminergic therapies — Used as monotherapies in some early stage patients but also often in conjunction with levodopa

- **MAO-B inhibitors: less potent, but better tolerated**
 - Inhibit dopamine metabolism
 - Limited by the amount of endogenous dopamine
 - Mild therapeutic benefit, but use may delay the introduction of levodopa for a few months to a year
 - Can also be used as add-on therapy to levodopa to reduce off-time
 - **Irreversible MAO-B inhibitors**
 - **Safer** — Not metabolized to amphetamine
 - PO QD: M Teva (Azilect/rasagiline)
 - TD patch QW: • Teikoku Pharma (TPU-002RA/rasagiline)
 - **Less safe** — Metabolized to small amounts of amphetamine
 - QD: • Teva/Valeant (Zelapar/orally disintegrating selegiline)
 - BID: G (selegiline)
 - **Reversible MAO-B inhibitors with additional dopaminergic activity** — Dual mechanism of action based on enhancement of dopaminergic function by reversible inhibition of MAO-B and inhibition of excessive glutamate release.
 - QD: M NDA Meiji/Newron/Zambon (Xadago/safinamide)
- **Dopamine agonists: more effective, but less tolerable**
 - Less efficacious than levodopa, but longer half-lives allow for more continuous dopaminergic stimulation
 - All have relatively similar efficacy.
 - Use may delay introduction of levodopa by several years.
 - Potentially dose-limiting side effects include somnolence, hallucinations, nausea, and low blood pressure.
 - **PO** — Choice of drug often comes down to tolerability, which varies by patient.
 - QD: G (Mirapex ER/pramipexole ER); G (Requip XL/ropinirole ER)
 - BID: G (bromocriptine mesylate)
 - TID: G (Mirapex/pramipexole); G (Requip/ropinirole)
 - Unknown: • Eli Lilly (undisclosed D1R agonist)* — *Agonist for the D1 dopamine receptor
 - **TD patches**
 - QD: M Otsuka/UCB (Neupro/rotigotine)
 - QW: O Corium (pramipexole or ropinirole)
- **Combination MAO-B inhibitor + dopamine agonist**
 - •• Pharma Two B (P2B001/rasagiline+pramipexole)

Adjunct therapies to basal levodopa

- **Modulate dopamine and glutamate release** — Adenosine A2a receptor antagonists
 - QD: RCT:1Q16 M ••• Kyowa Hakko Kirin (Nouriast/istradefylline)
 - BID: RCT:4Q17 ••• Acorda (SYN-115/tozadenant); •*/• Corvus/Vernalis (CPI-444) *being developed in immuno-oncology
 - Unknown dosing: RCT:YE18 •• Kyowa Hakko Kirin (KW-6356)
- **Alleviate 'off' time** — As the disease progresses and dopaminergic nerve terminals, degenerate, the concentration of dopamine becomes more dependent on plasma levodopa levels. Due to the short half-life of levodopa, 'off' episodes become more frequent.
 - **Short-acting dopaminergic rescue therapy** — For patients who experience unpredictable or unmanageable 'off' periods, these drugs bypass the GI tract to avoid the erratic absorption associated with oral levodopa.
 - **Inhaled levodopa**: NDA:2017 ••• Acorda (CVT-301/levodopa)
 - **Apomorphine** — This dopamine agonist initially requires antiemetic pretreatment to prevent nausea.
 - SC injection: M STADA/US WorldMeds (Apokyn/apomorphine)
 - Sublingual strip: RCT: 1Q17 ••• Sunovion (APL-130277/apomorphine)
 - Sublingual spray: O Renown (Apotone/apomorphine)
 - **Inhibit levodopa breakdown** → **Catechol-O-methyl transferase (COMT) inhibitors**
 - **No known risk of hepatotoxicity**
 - **Co-formulated with levodopa** — Replaces carbidopa+levodopa but commands a higher price: G (Stalevo/carbidopa+levodopa+entacapone)
 - **Taken with all doses of levodopa**: G (Comtan/entacapone)
 - **Not co-formulated**
 - TID: •• Orion (ODM-104)
 - QD: M Bial/Ono (Ongentys/opicapone)
 - **Risk of hepatotoxicity requires liver enzyme tests**
 - TID: G (Tasmar/tolcapone)
 - **Undisclosed Mechanism**: RCT: 3Q17 •• Pfizer (PF-06649751)
- **Treat levodopa-induced dyskinesia (LID)**
 - **Amantadine (multiple mechanisms)** — Antidyskinetic mechanism is unknown, but may be related to NMDA receptor antagonism.
 - QD: PDUFA:3Q17 NDA Adamas (Nurelin/amantadine ER); RCT: 1Q17 ••• Osmotica (Osmolex ER/amantadine ER)
 - BID/TID: G/OL (amantadine)
 - **Glutamate receptor modulators** — Block excessive corticostriatal glutamate signaling
 - **mGluR5 antagonist** — TID: •• Addex Therapeutics (ADX48621/dipraglurant-IR)
 - **NMDA receptor modulators** — BID: •• Otsuka (Nuedexta/dextromethorphan+quinidine); Unknown dosing: O Neurim (Neu-240)
 - **5-HT1A/B agonist** — Unknown mechanism; may block excessive corticostriatal glutamate signaling
 - BID: •• Amarantus/PsychoGenics (eltoprazine)

Invasive less convenient later-line therapies

Continuous subcutaneous infusion

- **Levodopa (better tolerated)** — Potential for improved efficacy and tolerability vs PO levodopa due to the steady levels of levodopa being delivered. Levodopa is unstable and technically challenging to concentrate. Patients who need large doses may require supplementation with PO levodopa.
 - **SC levodopa/carbidopa pump**
 - **Moderate PD** — One patch connected to a belt pump: RCT:4Q17 ••• NeuroDerm (ND0612L/carbidopa+levodopa)
 - **Severe PD** — Two patches connected to a belt pump: •• NeuroDerm (ND0612H/carbidopa+levodopa)
- **Other dopaminergic agonists (more AE considerations)** — Likely aimed at Parkinson's disease patients who do not respond well or cannot be treated with levodopa
 - **Fully internal device**
 - **SC rotigotine depot** — QW: • Serina Therapeutics (SER-214/rotigotine)
 - **Subdermal ropinirole implant** — Q3M-Q6M: O Titan Pharmaceuticals (ropinirole)
 - **External Pump, less convenient**
 - **SC apomorphine pumps**: •••/M STADA/US WorldMeds (APO-go PFS/apomorphine); • NeuroDerm (ND-0701/apomorphine)

Surgical procedures — These treatments are not usually considered until serious complications from levodopa arise, such as long periods of 'off time' and/or LID.

- **Gastrointestinal surgery** — Duopa is more expensive and generally less effective than deep brain stimulation (DBS) but has a different risk profile and can be used in patients with cognitive impairments.
 - **Pump continuously delivers levodopa into the duodenum**: M AbbVie (Duopa/carbidopa+levodopa gel); • NeuroDerm (ND0680/carbidopa+levodopa)
- **Neurosurgery** — Risks of serious or fatal AEs. Not considered for patients with cognitive impairment.
 - **Reversible and highly effective** — Efficacy can be as good as the patient's best response to levodopa, significantly improving motor scores and quality of life. DBS usually allows a decrease in levodopa dose, reducing side effects (including LID), but requires multiple follow-up visits to optimize therapy.
 - M (deep brain stimulation/DBS)
 - **Irreversible and unproven** — Potential one-time procedure with lasting therapeutic effect
 - **Gene therapy to boost dopamine production** — In PD patients, AADC is the rate-limiting enzyme for converting L-dopa to dopamine. Supplying AADC should allow for lower levodopa doses.
 - **AAV2 delivery of AADC**: • Sanofi/Voyager (VY-AADC01/AAV-hAADC)
 - **Lentiviral delivery of AADC+TH+CH1 to the striatum** — These 3 enzymes are needed for dopamine production, but may be poorly regulated when expressed by non-dopaminergic cells of the striatum.: •/•• Oxford BioMedica (ProSavin/OXB-102)

Manage non-motor symptoms

While PD is typically considered a movement disorder, patients often experience numerous non-motor symptoms including constipation, fatigue, sleep disorders, orthostatic hypotension, depression, psychosis and dementia. Many dopaminergic drugs exacerbate PD non-motor symptoms, which can further complicate treatment.

PD psychosis — Up to 40% of PD patients experience psychosis (hallucinations and/or delusions) at some point in their disease with ~10% being affected at any given time. Lowering dopaminergic drug dosage can improve psychotic symptoms, but exacerbates motor symptoms.

- **No sedation or effect on motor function** → **5-HT2A inverse agonist**: ACADIA (Nuplazid/pimavanserin) M; Arena Pharmaceuticals/Axovant (nelotanserin) O
- **Sedatives that may exacerbate motor symptoms** → **Atypical neuroleptics** — Boxed warning for increased mortality in elderly patients with dementia-related psychosis. Side effects include sedation, confusion, hypotension, and Parkinsonian motor symptoms.
 - **Effective, but weekly blood test for agranulocytosis required**: (clozapine) G/OL
 - **Less effective but much more commonly used because no blood testing is required**: (quetiapine) G/OL

PD dementia — 50-80% of patients with advanced PD will experience dementia.

- **Acetylcholinesterase inhibitors**
 - **Well-tolerated** → **TD patch**: (Exelon Patch/rivastigmine) G
 - **Poorly tolerated due to GI AEs** → **PO**: (donepezil) G/OL; (rivastigmine) G/OL
- **NMDA receptor antagonist**: (memantine) G/OL
- **Dual 5-HT2a/5-HT6 antagonist**: Acorda (SYN-120) •• RCT:2H16
- **5-HT2a inverse agonist**: Arena Pharmaceuticals/Axovant (nelotanserin) ••*/O *in Lewy Body Dementia
- **Undisclosed mechanism**: Eisai (E2027) •

Constipation and delayed gastric emptying — 50-90% of PD patients suffer from GI symptoms, which cause discomfort as well as variability of response to levodopa.

- **PO** → **Motilin receptor agonist**: GSK (GSK962040/camicinal) ••

Orthostatic Hypotension — Affects 30-40% of PD patients.

- **Norepinephrine prodrug** — Specifically approved for PD-related orthostatic hypotension. Also approved for freezing of gait in Japan. Being tested in phase 2 for PD dementia.: Lundbeck (Northera/droxidopa) M
- **Corticosteroid**: (fludrocortisone) G
- **Alpha-1 adrenergic receptor agonist**: (midodrine) G

Objective: Neuroprotection or Regeneration

There are no therapies proven to prevent or slow the progression of PD. Motor symptoms do not appear until a majority of dopaminergic neurons in the substantia nigra have died, suggesting that a neuroprotective therapy would ideally be administered prior to motor symptom onset. However, early biomarkers have not yet been validated and the pathogenesis of PD is not fully understood, increasing the difficulty and risk of pursuing neuroprotective strategies.

Target α-synuclein aggregates

α-synuclein is one of the primary constituents of Lewy bodies, the proteinaceous aggregates found in the neurons of PD patients. The identity of the toxic α-synuclein species is still unclear, but evidence favors soluble oligomers, with Lewy bodies potentially representing a protective aggregation. Although Lewy bodies are intracellular, toxic α-synuclein protofibrils may spread between neurons, contributing to disease progression.

IV: immunotherapy

- **Monoclonal antibodies recognizing α-synuclein**: Prothena/Roche (PRX002) ••*; Biogen/Neurimmune (BIIB054) • RCT: 2H17; AbbVie/BioArctic (BAN0805) O; Lundbeck O — *initiating in 2017
- **α-synuclein vaccine**: AFFiRiS (PD01A) •; AFFiRiS (PD03A) •
- **Antibody-like molecule** → **Bacteriophage protein that binds misfolded β sheets fused to human IgG-Fc**: NeuroPhage (NPT-088) O

PO: small molecules

- **Stabilize α-synuclein to prevent oligomerization**: Neuropore/UCB (NPT200-11) •
- **Increase α-synuclein clearance through the ubiquitin proteasome system (UPS)**: Oxalys (OXD-5) O
- **Target α-synuclein interaction with glucocerebrosides** — Lower levels of glucocerebrosidase are associated with more aggregated α-synuclein. Moreover, mutations in the glucocerebrosidase gene represent the greatest genetic risk factor for PD.
 - **Promote glucosylceramide breakdown** → **Glucocerebrosidase activators**: Amicus (AT3375) O; Allergan/Lysosomal Therapeutics (LTI291) O
 - **Reduce glucosylceramide production**
 - **Glucosylceramide synthase inhibitor**: Sanofi (GZ402671) ••
 - **Multidrug resistance glycoprotein-1 (MDR1) inhibitor**: Neuraltus (NP003) O
- **Target DJ-1 (PARK7) to inhibit α-synuclein aggregation** — Mutations in PARK7 cause autosomal recessive early onset PD
 - **Cell penetration DJ-1 protein**: Cantabio (CB201) O
 - **DJ-1 chaperone**: Cantabio (CB101) O; Cantabio (CB102) O

Intrathecal: targeted oligonucleotides

- **Prevent α-synuclein production** → **Oligonucleotides linked to a small molecule for targeted delivery** — siRNA or antisense-mediated knockdown of α-synuclein specifically within dopaminergic neurons: nLife Therapeutics (NLF-PD-1233) O

Protect neurons via anti-inflammatory or other mechanisms

PO: small molecules

- **Repurposed drugs**
 - Funded by non-profit and government sources.
 - Being tested in early-stage patients for potential to slow disease progression
 - **Calcium channel blocker** — Neurons that die from PD rely on a subtype of calcium channel to provide their steady pacemaking function, which may make them more vulnerable to insult and stress. Blocking this channel may be neuroprotective.: NIH (isradipine) •••
 - **Antioxidants**
 - **Urate precursor** — Higher levels of urate, a natural metabolite, have been associated with reduced risk of PD and slower rate of progression.: Michael J. Fox Foundation (inosine) ••
 - **Glutathione** — Reduced brain glutathione levels are associated with PD.: Michael J. Fox Foundation (intranasal glutathione) ••
 - **GLP-1 receptor agonist** — Approved for type 2 diabetes; Neuroprotective in some preclinical models: Michael J. Fox Foundation/UCL (Byetta/exenatide) ••
- **New chemical entities**
 - **Anti-inflammatory mechanism**
 - **Nurr1 activator** — Nurr1 is a transcription factor involved in establishing the fate of dopaminergic neurons and implicated in protection from neuroinflammation.: Sanofi (SA00025) O
 - **Myeloperoxidase (MPO) inhibitor** — MPO produces oxidants during inflammation, which can damage neurons. Also being tested in multiple system atrophy (MSA), another synucleinopathy: AstraZeneca (AZD3241) ••
 - **Unclear neuroprotective mechanism**
 - **JNK inhibitor** — JNK inhibition has been shown to be neuroprotective in several preclinical models.: Opko (SR-3306) O
 - **Nuclear retinoid acid receptor (RXR) agonist** — Several RXR activators have been shown to protect dopaminergic neurons from stress in preclinical models. This agonist has already been tested for a variety of cancers.: Allergan/Io Therapeutics (IRX4204) •
 - **Leucine-rich repeat kinase 2 (LRRK2) inhibitors** — On-target lung toxicity observed in animal models may be limiting. Gain-of-function mutations in the catalytic domains of LRRK2 are the most common genetic cause of PD, but the precise mechanism of neuropathology is unclear.
 - **Therapeutic window in preclinical models**: Pfizer (PF-06447475) O; Merck (MLi-2) O
 - **No therapeutic window in preclinical models**: Roche (GNE-7915) O
 - **Preclinical data unknown**: Arrien (ARN-1104) O; Ipsen/Oncodesign (ODS2005294) O; Denali O
 - **BCR-Abl inhibitors**
 - **Uncompelling phase 1 data** — Approved for CML: Georgetown (Tasigna/nilotinib) ••
 - **No data in PD to date** — In Ph1/2 for CML, targets Abl T315I resistance mutation: Sun Pharma (K-0706) •• RCT:4Q17
 - **Reversible redox co-factor**: Edison Pharma/Dainippon Sumitomo (EPI-589) O
 - **Hepatocyte growth factor mimetic**: M3 Biotechnology (MM201) O
 - **Reactive oxygen species (ROS) scavenger**: Aeolus (AEOL11114) O

TD

- **Nicotine patch** — Smoking is associated with a decreased risk of developing PD, but a recent study suggests that prodromal PD symptoms may just make it easier to quit.: Michael J. Fox Foundation (nicotine patch) ••

SC or IV

- **Recombinant neuregulin-1-b-1 peptide** — Preclinical evidence of dopaminergic neurotrophism and neuroprotection: Minerva Neurosciences (MIN-301) O
- **Recombinant IgG fusion proteins** — Monoclonal human insulin or transferrin receptor fused to therapeutic protein; Cross the BBB via receptor-mediated transport: ArmaGen (AGT-110/TNF decoy) O

Deliver neurotrophins to promote cell growth and survival

Highly invasive

- **One-time intracranial injection** → **AAV2-delivered glial-derived neurotrophic factor (GDNF)** — GDNF may support growth and survival of dopaminergic neurons.: NIH/uniQure (AAV2-GDNF) •
- **Chronic intracranial dosing**
 - **Intracranial injections of recombinant GDNF through indwelling catheter** — Supplied directly to the putamen. Previous attempts at delivering bolus GDNF into the ventricles failed to demonstrate a benefit and caused severe nausea/anorexia at higher doses, but targeted delivery to the striatum does not appear to cause the same side effects.: MedGenesis/Amgen (liatermin/GDNF) FAILED
 - **Intraventricular pump: platelet-derived growth factor (PDGF-BB)** — PDGF may stimulate neurogenesis along the ventricular wall. Continuous delivery using implanted catheter and infusion pump (Medtronic): Newron (sNN0031) FAILED

Less invasive

- **Intranasal** → **Intranasal administration: GDNF non-viral DNA nanoparticles** — Compacts single copies of DNA into nanoparticles that can be taken into the cell by membrane-associated proteins: Copernicus Therapeutics (GDNF nanoparticles) O
- **Likely IV** → **Recombinant IgG fusion proteins** — Monoclonal human insulin or transferrin receptor fused to therapeutic protein; Cross the BBB via receptor-mediated transport: ArmaGen (AGT-190/GDNF) O; ArmaGen (AGT-115/EPO) O

Protect or replace neurons via intracranial cell transplantation

Restore dopamine production

- Human fetal dopamine neurons have provided long-term benefit in some patients but no benefit or troublesome dyskinesias in others.
- Two placebo-controlled trials failed to meet their primary endpoints.
- **Fetal neural precursor cells** — Multiple fetuses required per patient: TRANSEURO (fetal dopaminergic cells) •
- **Parthenogenetic stem cell-derived dopamine neuron precursors** — Unlimited cell source; Ability to generate authentic dopamine neurons is questionable: International Stem Cell •
- **Embryonic stem cell-derived dopamine neurons precursors** — Unlimited cell source; Most authentic dopamine neurons generated to date: Memorial Sloan Kettering Cancer Center O

Protect neurons — Early treatment likely required

- **Porcine-derived alginate-encapsulated choroid plexus cells**: Living Cell Technologies (NTCELL) ••
- **Encapsulated GDNF-secreting retinal pigmented epithelial cells**: NsGene (NsG0301) O
- **Human genetically modified mesenchymal stromal cells (MSCs)**
 - **Autologous**: BrainStorm Cell Therapeutics (NurOwn/MSC-NTF) ••*/O *in ALS
 - **Allogeneic**: SanBio (SB623/Notch-MSCs)/Sumitomo Dainippon O *in stroke

TECHATLAS PARKINSON'S DISEASE SOLUTIONS DEVELOPMENT

Company	Levodopa: Extended release	Levodopa: Continuous	Levodopa: Rescue	Dopamine agonist: Oral QD	Dopamine agonist: Transdermal QD	Dopamine agonist: Rescue	MAO-B inhibitor: Oral QD	MAO-B inhibitor: Transdermal QW	α-synuclein-targeted therapy: Immunotherapy	α-synuclein-targeted therapy: Small molecule	α-synuclein-targeted therapy: Oligonucleotide	Adenosine A2a antagonist	Amantadine
AbbVie		M											
Impax	M •												
Acorda			•••									•••	
Teva							M						
UCB					M					•			
Kyowa Hakko Kirin												•••	
Roche									•				
Biogen									•				
Lundbeck									•				
Acquirable assets	•• Intec	•/•• NeuroDerm		•• Pharma Two	Corium	••• Cynapsus	••• Newron	• Teikoku Pharma	• AFFiRiS; BioArctic; NeuroPhage	Oxalys	nLife	•/•• Vernalis	••• Adamas; ••• Osmotica

TECHATLAS MILESTONES: RCT DATES

Symptom treated	Class	Company (Drug)	RCT date
'Off' time	Adenosine A2a receptor antagonist	Kyowa Hakko Kirin (Nouriast/istradefylline)	1Q16 3
		Biotie (SYN115/tozadenant)	4Q17 3
	Sublingual apomorphine rescue therapy	Cynapsus (APL-130277/apomorphine)	2Q16 3
	Inhaled levodopa rescue therapy	Acorda (CVT-301/levodopa)	3Q16 3
Levodopa-induced dyskinesia	Amantadine	Adamas (Nurelin/amantadine ER)	1Q16 3
		Osmotica (Osmolex/amantadine ER)	2Q16 3
	NMDA receptor modulator	Otsuka (Nuedexta/dextromethorphan+quinidine)	4Q15 2
Disease progression	α-synuclein mAb	Prothena/Roche (PRX002)	1H16 1
		Biogen/Neurimmune (BIIB054)	2H16 1
PD psychosis	5-HT2a inverse agonist	ACADIA (Nuplazid/pimavanserin)	2Q16 P
PD dementia	5-HT2a/5-HT6 dual antagonist	Biotie (SYN-120)	2H16 2

Timeline: 2016 (Q1 Q2 Q3 Q4) | 2016 (Q1 Q2 Q3 Q4) | 2016 (Q1 Q2 Q3 Q4) | 2016 (Q1 Q2 Q3 Q4)

3 Phase 3 data
2 Phase 2 data
1 Phase 1 data
P PDUFA

Clinical stage codes

O	Preclinical/Discovery	⊘	Patent Expiry
•	Phase 1	PDUFA	Prescription Drug User Fee Act (deadline for FDA decision)
••	Phase 2	ANDA	Abbreviated New Drug Application
•••	Phase 3	OL	Off-label
M	Marketed	OTC	Over-the-Counter
G	Generic	NDA	New Drug Application
RCT	Randomized Controlled Trial	BLA	Biologics License Application
IST	Investigator-Sponsored Trial	MAA	Marketing Authorization Application (Europe's NDA)
UCT	Uncontrolled Trial	CP	Compounding Pharmacy
M/•••	Phase in US	DIS-CONTINUED	Discontinued/Terminated/Suspended
M/•••	Phase in EU or ex-US	FAILED	Failed

Routes of delivery

IV	Intravenous	IB	Intravesical/Intrabladder
ID	Intradermal	IC	Intracerebral
SL	Sublingual	TD	Transdermal
IM	Intramuscular	EC	Epicutaneous
SC	Subcutaneous	IN	Intranasal
PO	Oral	SGI	Salivary gland injection
IVT	Intravitreal		
ICM	Intracisternal		
ICV	Intracerebroventricular		
ITH	Intrathecal		
IT	Intratumoral		
TD	Transdermal		
IPL	Intrapleural		
IP	Intraperitoneal		

Dosage

QD	Once a day
BID	Twice a day
TID	Three times a day
QOD	Every other day
QW	Once a week
Q2W	Once every 2 weeks
QM	Once a month

GENERAL DISCLAIMERS:

The information contained herein (the "Materials") regarding RA Capital Management, L.P. and its affiliates and/or any investment products it advises (collectively, "RA Capital") is provided for informational and discussion purposes only and is not, and may not be relied on in any manner, as legal, tax, or investment advice. Each recipient should make its own investigations and evaluations of RA Capital, any investment products it advises, and the Materials, and should consult its own attorney, business adviser, and tax adviser as to legal, business, tax, and related matters concerning the Materials. The information contained in the Materials is not intended to be, and should not be viewed as, "investment advice" within the meaning of 29 C.F.R. §2510.3-21 or otherwise.

The Materials are not an offer or solicitation for the purchase or sale of any security including any interest in RA Capital Healthcare Fund, L.P. (the "Master Fund") or RA Capital Healthcare International Fund, Ltd. (the "Offshore Fund," and collectively with the Master Fund, the "Fund"), and should not be construed as such. Such an offer will only be made by means of a confidential Private Placement Memorandum (the "PPM") to be furnished to qualified investors upon request. The information contained herein is qualified in its entirety by reference to the PPM, which contains additional information about the investment objective, terms, and conditions of an investment in the Fund, and also contains certain disclosures that are important to consider when making an investment decision regarding the Fund. In the case of any inconsistency between any information contained herein and the PPM, the terms of the PPM shall control.

The Materials are proprietary and confidential and may include commercially sensitive information, must be kept strictly confidential, and may not be copied, used for an improper purpose, reproduced, republished, or posted in whole or in part, in any form, without the prior written consent of RA Capital. The recipient of the Materials must not make any communication regarding the information contained herein, including disclosing that the Materials have been provided to such recipient, to any person other than its authorized representatives assisting in considering the information contained herein. Each recipient agrees to the foregoing and to return (or destroy upon RA Capital's instructions) the Materials promptly upon request.

Any investment strategies discussed herein are speculative and involve a high degree of risk, including loss of capital. Investments in any products described herein and the Fund's performance can be volatile, and investors should have the financial ability and be willing to accept such risks. An investor could lose all or a substantial amount of his or her investment. The Fund may be leveraged. Interests in the Fund are illiquid, as there is no secondary market for the Fund interests, and none is expected to develop. The Fund interests are subject to restrictions on transfer. Prior to investing in the Fund, investors should read the PPM and pay particular attention to the risk factors contained therein. Fees and expenses charged in connection with an investment in the Fund may be higher than the fees and expenses of other investment alternatives and may offset investment profits of the Fund. RA Capital has total trading authority over the Fund. The use of a single advisor applying generally similar trading programs could mean lack of diversification and, consequentially, higher risk. A portion of the trades executed for the Fund may take place on foreign exchanges. It should not be assumed, and no representation is made, that past investment performance is reflective of future results. Nothing herein should be deemed to be a prediction or projection of future performance. To the extent any prior or existing investments are described, RA Capital makes no representations, and it should not be assumed, that past investment selection is necessarily reflective of future investment selection, that any performance discussed herein will be achieved or that similar investment opportunities will be available in the future or, if made, will achieve similar results.

In particular, to the extent valuation information is provided for any unrealized investments, such valuations are RA Capital's estimates as of the date set forth in the Materials, and there can be no assurance that unrealized investments will be realized at such valuations. While RA Capital believes any valuations presented herein are reasonable, such valuations may be highly subjective, particularly for private investments, are based on information provided by third parties and/or RA Capital's assumptions, any or all of which might be mistaken or incomplete. Actual realized returns will depend on, among other factors, future operating results, the value of the assets and market conditions at the time of disposition, any related transaction costs, and the timing and manner of sale, all of which may differ from the assumptions on which the valuations contained herein are based. As a result of the foregoing, actual realized returns may differ materially from the valuations contained herein.

References, either general or specific, to securities and/or issuers are for illustrative purposes only and are not intended to be, and should not be interpreted as, recommendations to purchase or sell such securities. Certain current and prior investments may be highlighted in order to provide additional information regarding RA Capital's investment strategy, the types of investments it pursues, and anticipated exit strategies. In addition, due to confidentiality restrictions, the information contained herein might not reference investments in certain companies. Certain information contained herein concerning economic trends and/ or data is based on or derived from information provided by independent third-party sources. RA Capital believes that the sources from which such information has been obtained are reliable; however, it cannot guarantee the accuracy of such information and has not independently verified the accuracy or completeness of such information or the assumptions on which such information is based.

The Materials contain statements of opinion and belief. Any views expressed herein, unless otherwise indicated, are those of RA Capital as of the date indicated, are based on information available to RA Capital as of such date, and are subject to change, without notice, based on market and other conditions. No representation is made or assurance given that such views are correct. RA Capital has no duty or obligation to update the information contained herein.

Certain information contained in this document constitutes "forward-looking statements," which can be identified by the use of forward-looking terminology such as "may," "will," "should," "expect," "anticipate," "target," "project," "estimate," "intend," "continue," or "believe," or the negatives thereof or other variations thereon or comparable terminology. Due to various risks and uncertainties, actual events or results or the actual performance of any investment may differ from those reflected or contemplated in such forward-looking statements. Prospective investors should not rely on these forward-looking statements when making an investment decision.

None of the information contained herein has been filed with the U.S. Securities and Exchange Commission, any securities administrator under any securities laws of any U.S. or non-U.S. jurisdiction, or any other U.S. or non-U.S. governmental or self-regulatory authority. No such governmental or self-regulatory authority will pass on the merits of any offering of interests by RA Capital or the adequacy of the information contained herein. Any representation to the contrary is unlawful. The Fund interests have not been and will not be registered under the U.S. Securities Act of 1933, as amended, or qualified or registered under any applicable state, local, provincial, or other statutes, rules, or regulations. The Fund has not been, and will not be, registered as an investment company under the U.S. Investment Company Act of 1940, as amended.